PKPD Modeling of Brain Receptor Occupancy from Animal to Man for Establishing the Clinical Dose of the NK-1 Antagonist LY686017

Christine Falcoz [1], Wenping Wang [2], William Ebling [3], Laurent Claret [1], Smriti Iyengar [4], Donald R. Gehlert [4], William Kielbasa [4]

[1] Pharsight Corporation, Mountain View, CA, USA, [2] now at deCODE genetics, Reykjavik, Iceland, [3] now at Emergent Insights Consulting Inc, Newark, DE, USA, [4] Eli Lilly and Company, Indianapolis, IN, USA.

ABSTRACT

Background: LY686017 is a neurokinin-1 (NK-1) antagonist that has promise as a potential novel treatment for alcoholism. Mice deficient in NK-1 showed a voluntary decrease in consumption and increased sensitivity to the sedative effects of alcohol. LY686017 showed activity in gerbil models of anxiety at brain NK-1 receptor occupancy (RO) levels of 80% or greater. The clinical development of LY686017 included a human positron emission tomography (PET) study to determine brain NK-1 RO to optimize clinical dose selection before assessing the potential efficacy of LY686017 in proof of concept (POC) studies. The gerbil model was used to help predict clinical doses below and at the target level of at least 80% RO.

Aim: Simulate trough RO in humans to optimize and confirm dose selection prior to the RO PET Phase I study in healthy subjects and, later, prior to POC Phase 2 studies in patients.

Methods: Models were developed sequentially to guide the design of human studies. Step 1: In gerbil, PK (0.2-5 mg/kg) and PKRO (0.01-1 mg/kg) models were built following single dosing. Step 2: A preliminary human PK model was developed from single dose data (1-800 mg) and along with estimate of gerbil EC50, simulation of trough human RO was used to select doses to characterize the dose-RO curve in the planned PET study. Step 3: Using the PET study results at trough during steady state (1–100 mg QD), the PK model was refined and a PKRO model was developed in humans. Step 4: RO was simulated to confirm the LY686017 dose for POC studies.

Results: Step 1: The gerbil EC50 was determined at 0.7 ng/mL. When normalized for plasma binding, the predicted human EC50 was 2.8 ng/mL. Step 2: The first human PK model included dose-dependent first-order absorption. Simulations of trough RO showed that a 1 mg dose was needed to characterize the dose - RO relationship. Step 3: The mean observed NK-1 RO was 25, 75, 80 and 93% at 1, 10, 30 and 100 mg QD, respectively. Observed trough concentrations and RO values were well predicted in Step 2, except for an under prediction at 1 mg, and the PK model was refined with two absorption processes to adequately represent the measured trough concentrations. The EC50 for RO in humans was about 3 ng/mL. Step 4: RO simulations with the human PKRO model confirmed the dose of 50 mg QD for POC studies: RO predictions were > 90% as a mean and > 85% in at least 90% of subjects. Step 5: The POC study in social anxiety disorder did not show significant efficacy; however, plasma concentrations indicated receptor saturation based on the PKRO model. In the POC study for treatment of alcoholism, LY686017 suppressed spontaneous alcohol cravings and improved overall well-being in recently detoxified alcoholic patients.

Conclusion: The human EC50 for RO was predicted from the gerbil model, validating the species as a preclinical surrogate of clinical RO for an NK-1 receptor antagonist. The RO time course in humans was predicted early in development using the gerbil PKRO model and the human PK model. The PKPD modeling and simulation framework from animal to human led to optimal dosing decisions. Clinical doses > 50 mg are unlikely to lead to improved exposure or more extensive RO. Further evaluation of LY686017 for the treatment of alcoholism is warranted.

BACKGROUND

Substance P and Antagonists

- Substance P belongs to the tachykinin family of peptide neurotransmitters
- NK-1 is a G-protein coupled receptor (GPCR) for which Substance P is an agonist
- Substance P and NK-1 receptors are expressed in brain areas involved in stress responses and drug reward
- Potential for treatment of pain, depression, anxiety, alcoholism
- Clinical efficacy data exist for NK-1 antagonists in the treatment of depression, anxiety and to prevent chemotherapy-induced nausea and vomiting (CINV).

Preclinical Models

- Mice deficient in NK-1 showed a voluntary decrease in consumption and increased sensitivity to the sedative effects of alcohol, compared to heterozygous and wild-type mice
- In gerbil, maximal anti-anxiety effects (foot taping (FT)) were observed at > 80% receptor occupancy (RO).

Fig. 1: Dose Response for Effect and RO in Gerbil

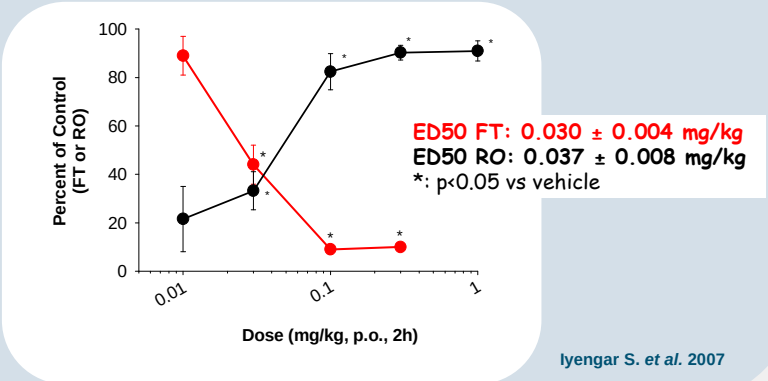

Percent of Control (FT or RO)
ED50 FT: 0.030 ± 0.004 mg/kg
ED50 RO: 0.037 ± 0.008 mg/kg
*: p<0.05 vs vehicle
Dose (mg/kg, p.o., 2h)

Iyengar S. *et al.* 2007

Clinical Development of LY686017

Several questions were to be addressed

- Is LY686017 a brain-permeable molecule in humans?
- What range of doses should be tested to characterize the dose-RO relationship?
- What dose has significant target blockade (high RO)?
- Can RO be used as a surrogate for efficacy in the clinic?

Three studies were planned

- A single-dose ranging study to assess safety and PK
- A multiple-dose ranging study to assess safety, PK and RO using positron emission tomography (PET)
- Proof-of-concept (POC) studies in social anxiety disorder (SAD) and alcohol dependence.

METHODS

A sequential model development guided the design of human studies

Step 1: Gerbil PK and PKRO models after single dose

- PK (0.2-5 mg/kg)
- PKRO (0.01-1 mg/kg) using RO data obtained at 2 h (~ Cmax)

Step 2: Design of the PET study in man

- Preliminary human PK model (1-800 mg single doses)
- Preliminary human PKRO model leveraging the gerbil EC50
- Simulate clinical doses that have optimal target engagement and best characterize the dose/concentration-RO relationship.

Step 3: Improved PK and final PKRO models in man

- Using the PET study results (1–100 mg QD)

Step 4: Dose selection for POC studies

- Simulate RO to confirm the LY686017 dose for POC studies.

Model Building Assumptions

- Only LY686017 (parent) was responsible for receptor blockade
- The PKRO relationship was direct
- Gerbil and human PKRO relationship is identical following correction for plasma protein binding
- Human single dose PK can predict multiple dose PK.

Pharsight

Lilly Answers That Matter

RESULTS

Step 1: Model Development in Gerbil

Fig. 2: PK, RO and PKRO Data in Gerbil

PK data	**RO data vs. dose**	**PKRO data**
0.2, 1, 5 mg/kg → 2 compartment model with 1st order absorption	0.01, 0.03, 0.1, 0.3, 1 mg/kg	→ Emax model EC50 = 0.7 ng/mL ROmax = 91%

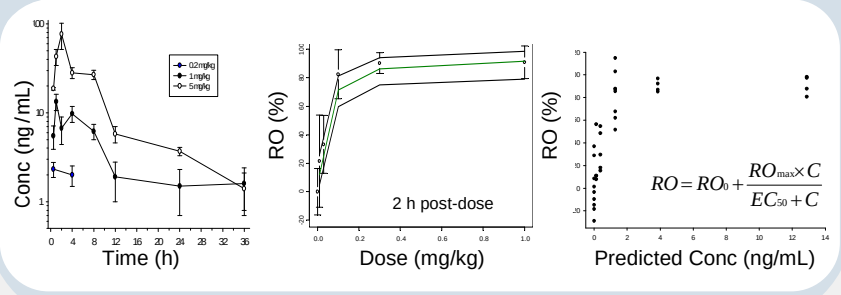

Conc (ng/mL)
Time (h)
0.2mg/kg
1mg/kg
5mg/kg
RO (%)
2 h post-dose
Dose (mg/kg)
RO (%)
$RO = RO_0 + \frac{RO_{max} \times C}{EC_{50} + C}$
Predicted Conc (ng/mL)

Step 2: Design of the Human PET Study

The preliminary human PKRO model utilized:

1) A first PK model in man. PK data from the single-dose study showed a nonlinearity in drug exposure, attributed to absorption (dissolution limited).
2) The gerbil EC50. After accounting for differences in plasma protein binding between species, the human EC50 was predicted at 2.8 ng/mL.

Fig. 3: Preliminary PKRO Model in Human

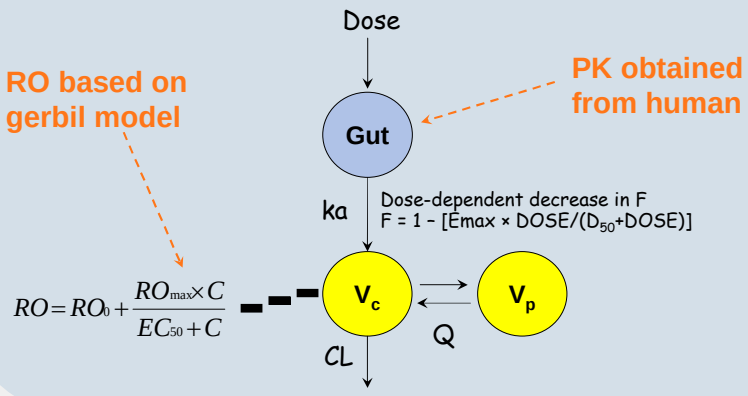

Dose
RO based on gerbil model
PK obtained from human
Gut
ka
Dose-dependent decrease in F
$F = 1 - [E_{max} \times DOSE/(D_{50}+DOSE)]$
$RO = RO_0 + \frac{RO_{max} \times C}{EC_{50} + C}$
V_c
V_p
Q
CL

Simulations of concentrations and RO were performed using the design of the PET study

Design of the PET study

- 28-day exposure, 4 dose levels QD, weekly dose escalation
- PET scan after 7th day of dosing at trough for each dose
- 3 PET scans per subject (baseline and 2 post-dose)
- Possible dose range 1–800 mg QD.

A 10 mg dose was predicted to achieve >80% RO at trough. A lower dose was desirable to characterize dose dependence in RO.

Fig. 4: Simulation Using the PET Study Design

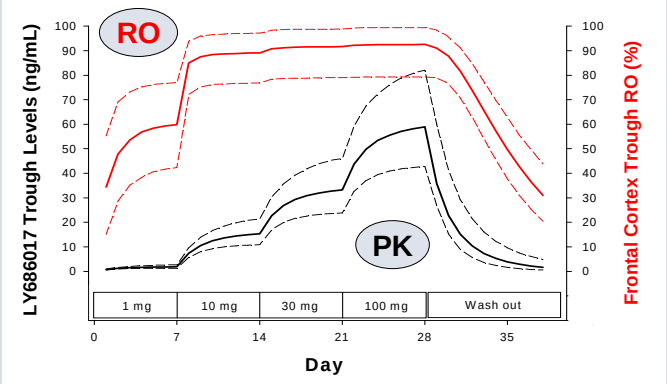

RO
PK
LY686017 Trough Levels (ng/mL)
Frontal Cortex Trough RO (%)
1 mg
10 mg
30 mg
100 mg
Wash out
Day

Step 3: Improvement of the PK Model and Final PKRO Model in Man

The results of the PET study confirmed prior simulations, excepted an under prediction at 1 mg.

Fig. 5: Results vs. Prior Simulations for the PET Study

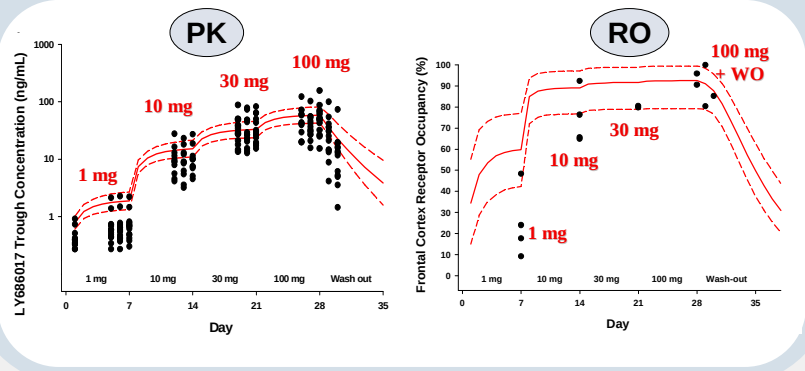

PK
RO
LY686017 Trough Concentration (ng/mL)
1 mg
10 mg
30 mg
100 mg
Wash out
Day
Frontal Cortex Receptor Occupancy (%)
100 mg + WO
Wash-out

The relationship was direct between RO and trough concentrations

- At 100 mg, the relationship was the same during dosing & washout
- EC50 estimated from observed data was 2.5 ng/mL (Emax=98%).

Fig. 6: RO vs. Trough Levels Observed in the PET Study

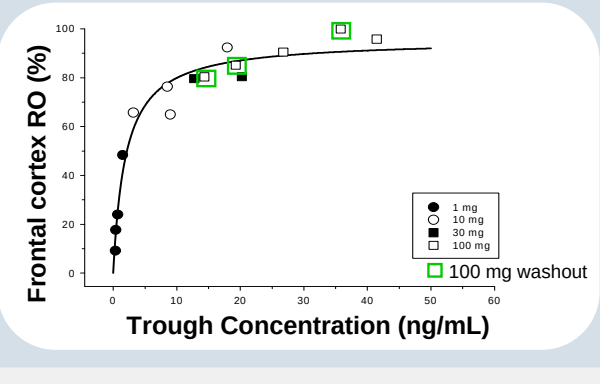

Frontal cortex RO (%)
1 mg
10 mg
30 mg
100 mg
100 mg washout
Trough Concentration (ng/mL)

The PK model required the addition of a zero-order absorption process to adequately characterize trough concentrations at steady-state.

Fig. 7: Improved Human PK Model

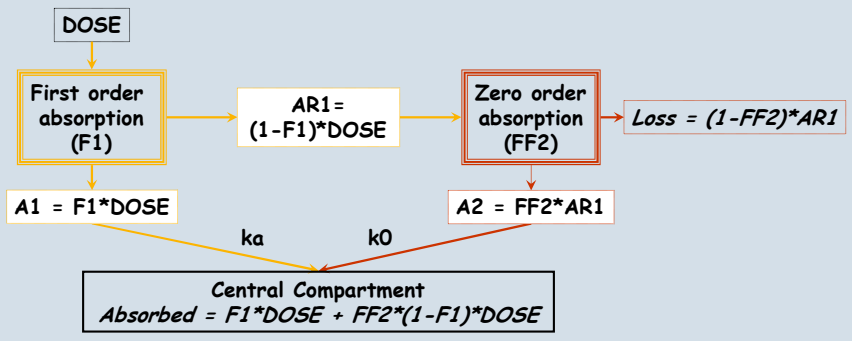

DOSE
First order absorption (F1)
AR1= (1-F1)*DOSE
Zero order absorption (FF2)
Loss = (1-FF2)*AR1
A1 = F1*DOSE
A2 = FF2*AR1
ka
k0
Central Compartment
Absorbed = F1*DOSE + FF2*(1-F1)*DOSE

F1 = 1 - (Dose/(D50 + Dose))
First order absorption
Emax=1

FF2 = 1 - (AR1/(A50+AR1))
Zero-order absorption
Emax=1

F2 = (1 - F1) × F F2

F = F1 + F2
Relative oral bioavailability

	Parameter	Estimate	SE
$\theta 1$	CL/F	23	2.2
$\theta 2$	V2=Vc/F	100	6.6
$\theta 3$	V3=Vp/F	334	30
$\theta 4$	Q/F	17	2.1
$\theta 5$	KA	3.2	0.88
$\theta 6$	D50_F1	6.5	0.78
$\theta 7$	A50_FF2	34	5.47
$\theta 8$	D2 (duration of zero-order process)	56	0.49
$\theta 9$	Error: additive	0.46	0.058
$\theta 10$	Error: proportional	0.55	0.020
$\eta 1$	eta_CL/F	0.12	0.043
$\eta 2$	eta_V2	0.083	0.025
$\eta 3$	eta_A50_F2	0.44	0.14
$\varepsilon 1$	Proportional	0.049	0.007

An EC50 of 2.8 ng/mL was estimated for the final PKRO model in man, using observed RO data and trough concentrations predicted with the PK model.

Fig. 8: Prediction of Trough RO at Steady State (vs. Obs)

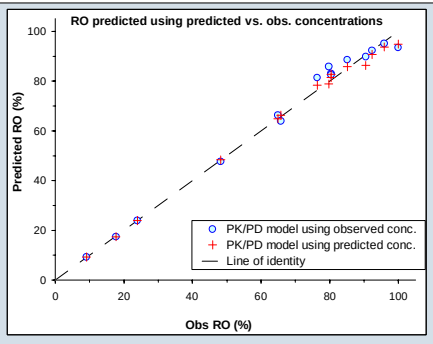

RO predicted using predicted vs. obs. concentrations
Predicted RO (%)
PK/PD model using observed conc.
PK/PD model using predicted conc.
Line of identity
Obs RO (%)

Step 4: Dose Selection for POC Studies

- The final human PKRO model was used to confirm the dose for POC studies
- Simulations showed that exposure saturated at ≥50 mg
- The dose selected was 50 mg. RO was predicted to exceed 90% at trough following once daily dosing.

Tab. 1: Dose Selection for POC Studies

Dose (mg QD)			1	10	20	30	50	100
Trough level (ng/mL)	Simulated	Median	0.74	10	19	25	33	42
		10% quantile	0.36	6.2	11	14	18	21
		90% quantile	1.6	19	33	44	59	80
	Observed	Mean	0.59	9.1	-	28	-	43
RO at trough (%)	Simulated	Median	21	79	87	90	92	94
		10% quantile	11	69	80	84	86	88
		90% quantile	37	87	92	94	96	97
	Observed	Mean	25	75	-	80	-	93

Simulations: after 12 weeks of treatment, number of subjects = 74, number of replicates = 500
Doses used in the PET study were 1, 10, 30 and 100 mg QD

Fig. 9: Simulations of the Dose influence on Trough Levels and RO at Steady State

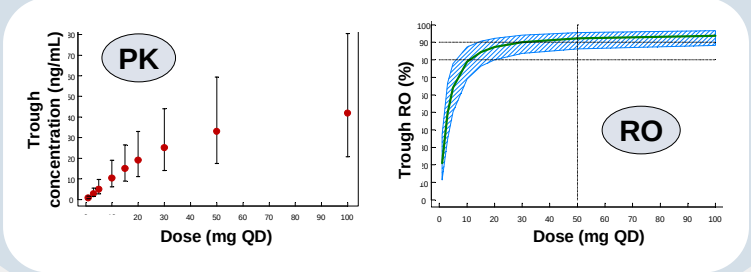

PK
Trough concentration (ng/mL)
Dose (mg QD)
RO
Trough RO (%)
Dose (mg QD)

Step 5: Clinical Efficacy Outcomes in POC Studies

POC study in social anxiety disorder

- LY686017 did not show significant efficacy
- LY686017 plasma concentrations indicated that saturation of NK-1 receptors would be expected during the dosing interval, based on PKRO modeling data.

POC study for treatment of alcoholism

- LY686017 suppressed spontaneous alcohol cravings, improved overall well-being, blunted cravings induced by a challenge procedure, and attenuated concomitant cortisol responses
- Brain fMRI responses to affective stimuli likewise suggested beneficial LY686017 effects
- As assessed by these surrogate markers of efficacy, LY686017 warrants further investigation as a treatment target in alcoholism.

CONCLUSION

- The human PKRO model was clinically predictive based on PET results and validated the gerbil as an animal model for clinical RO for the NK-1 antagonist (Platform)
- The RO time course in humans was predicted early in development using the gerbil PKRO model and a first human PK model
- PKRO modeling revealed that QD dosing of >30 mg LY686017 leads to sustained trough RO at 80% or greater with 90% confidence, and that doses of more than 50 mg are unlikely to lead to improved exposure or more extensive RO
- The PKPD modeling and simulation framework from animal to human directly supported optimal dosing decisions
- A single dose strength of 50 mg was chosen for POC studies and supported a quick win/quick kill strategy
 - Negative POC… quick kill without asking “Did we get the dose right?”. The mechanism does not appear to be suitable for indication.
 - Positive POC… quick win. Integrate model to understand the relationship between RO and clinical efficacy in definitive studies.
- Further evaluation of LY686017 for the treatment of alcoholism is warranted.

PKUK 2008